Impact of oral ferric maltol and IV iron on health-related quality of life in patients with iron deficiency anaemia and inflammatory bowel disease, and relationship with haemoglobin and serum iron

P567

Stefanie Howaldt[1], Ian Jacob[2], Mark Sampson[3], Fatma Akriche[4]

[1]MZV für Immunologie, Hamburg, Germany; [2]Health Economics and Outcomes Research Ltd, Cardiff, United Kingdom; [3]Shield Therapeutics PLC, London, United Kingdom; [4]Norgine Ltd, Rueil Malmaison, France.

Background

Iron deficiency anaemia (IDA) is common in patients with inflammatory bowel disease (IBD). Symptoms include fatigue, dyspnoea, palpitations and headache, and can significantly impair health-related quality of life (HRQoL) over and above the impact of the underlying disease.[1,2]

IV iron is currently the main treatment for patients intolerant or unsuitable for standard oral iron.[3] Due to the risk of hypersensitivity reactions, it must be administered in a setting with full resuscitation facilities.[4] Intolerance to oral iron is common due to oxidation of unabsorbed ferrous (Fe^{2+}) iron and consequent damage to the gastrointestinal tract from reactive oxygen species.[5,6] Ferric maltol (FM), a stable oral complex of ferric (Fe^{3+}) iron and maltol, is designed to provide efficient iron delivery and minimise formation of free iron in the gut, thus reducing the potential for gastric adverse events.[6] It is approved in the EU for the treatment of iron deficiency in adults.[7]

The HRQoL benefits of FM and IV ferric carboxymaltose (FCM) and their relationship to haematological parameters were evaluated in an exploratory post hoc analysis using data from a randomised controlled trial (NCT02680756).[8]

Trial design and primary findings

Patients with IBD and IDA (haemoglobin [Hb] ≥8.0 g/dL and ≤11.0 g/dL for women or ≥8.0 g/dL and ≤12.0 g/dL for men, and ferritin <30 ng/mL or ferritin <100 ng/mL with transferrin saturation <20%) were randomised to FM (30 mg b.d.) or IV FCM (as per local Summary of Product Characteristics or prescribing information) in an open-label, Phase 3b non-inferiority study. The primary endpoint was Hb responder rate (proportion of patients achieving a ≥2 g/dL increase or normalisation of Hb at week 12). HRQoL was assessed via the Short Form Health Survey (SF-36).

- 250 patients were randomised: 125 to FM (per-protocol [PP] n=86) and 125 to IV FCM (PP n=92).
- The Hb responder rate for oral FM was non-inferior to IV FCM in the PP population (74% vs 83%); risk difference was -0.1 (two-sided p=0.017; 95% CI -0.2, 0.0), within the pre-defined non-inferiority margin of 20% difference.

Methods

Analysis of HRQoL and haematological parameters

In a post hoc analysis of patient-level data, Hb, serum iron, serum ferritin and SF-36 scores (physical component summary [PCS], mental component summary [MCS] and individual domain scores) were analysed at baseline and week 12.

Correlations between SF-36 scores (PCS and MCS) and haematological parameters were assessed via Pearson's correlation coefficient (PCC) using pooled data from the FM and IV FCM treatment arms. Change from baseline in SF-36 scores and correlations between parameters were summarised graphically and descriptively. SF-36 domain score data was included for all patients who had data within the required time windows (FM: N=109; IV FCM: N=114).

Results

HRQoL and clinical parameter improvements

- Hb, serum iron, serum ferritin and HRQoL all improved with both treatments at Week 12.
- Improvements in SF-36 PCS and MCS scores were slightly greater with FM than with IV FCM (difference not statistically significant; Table 1).
- Scores improved across all SF-36 domains for both FM and IV FCM (Figure 1).

Correlations between HRQoL and clinical parameters

Improvements in Hb and serum iron were positively associated with improvements in HRQoL (PCS and MCS; Table 2; Figure 2).

Serum ferritin improved between baseline and week 12, but no clear association was found with changes in HRQoL (PCC = -0.075 for PCS and -0.013 for MCS).

Table 1: Change in PCS and MCS scores from baseline to week 12

	PCS		MCS	
	Change	p-value*	Change	p-value*
Oral FM	+3.23	0.22	+3.89	0.27
IV FCM	+2.09		+2.52	

*FM vs. IV FCM

Figure 1: Mean SF-36 domain scores at baseline and week 12

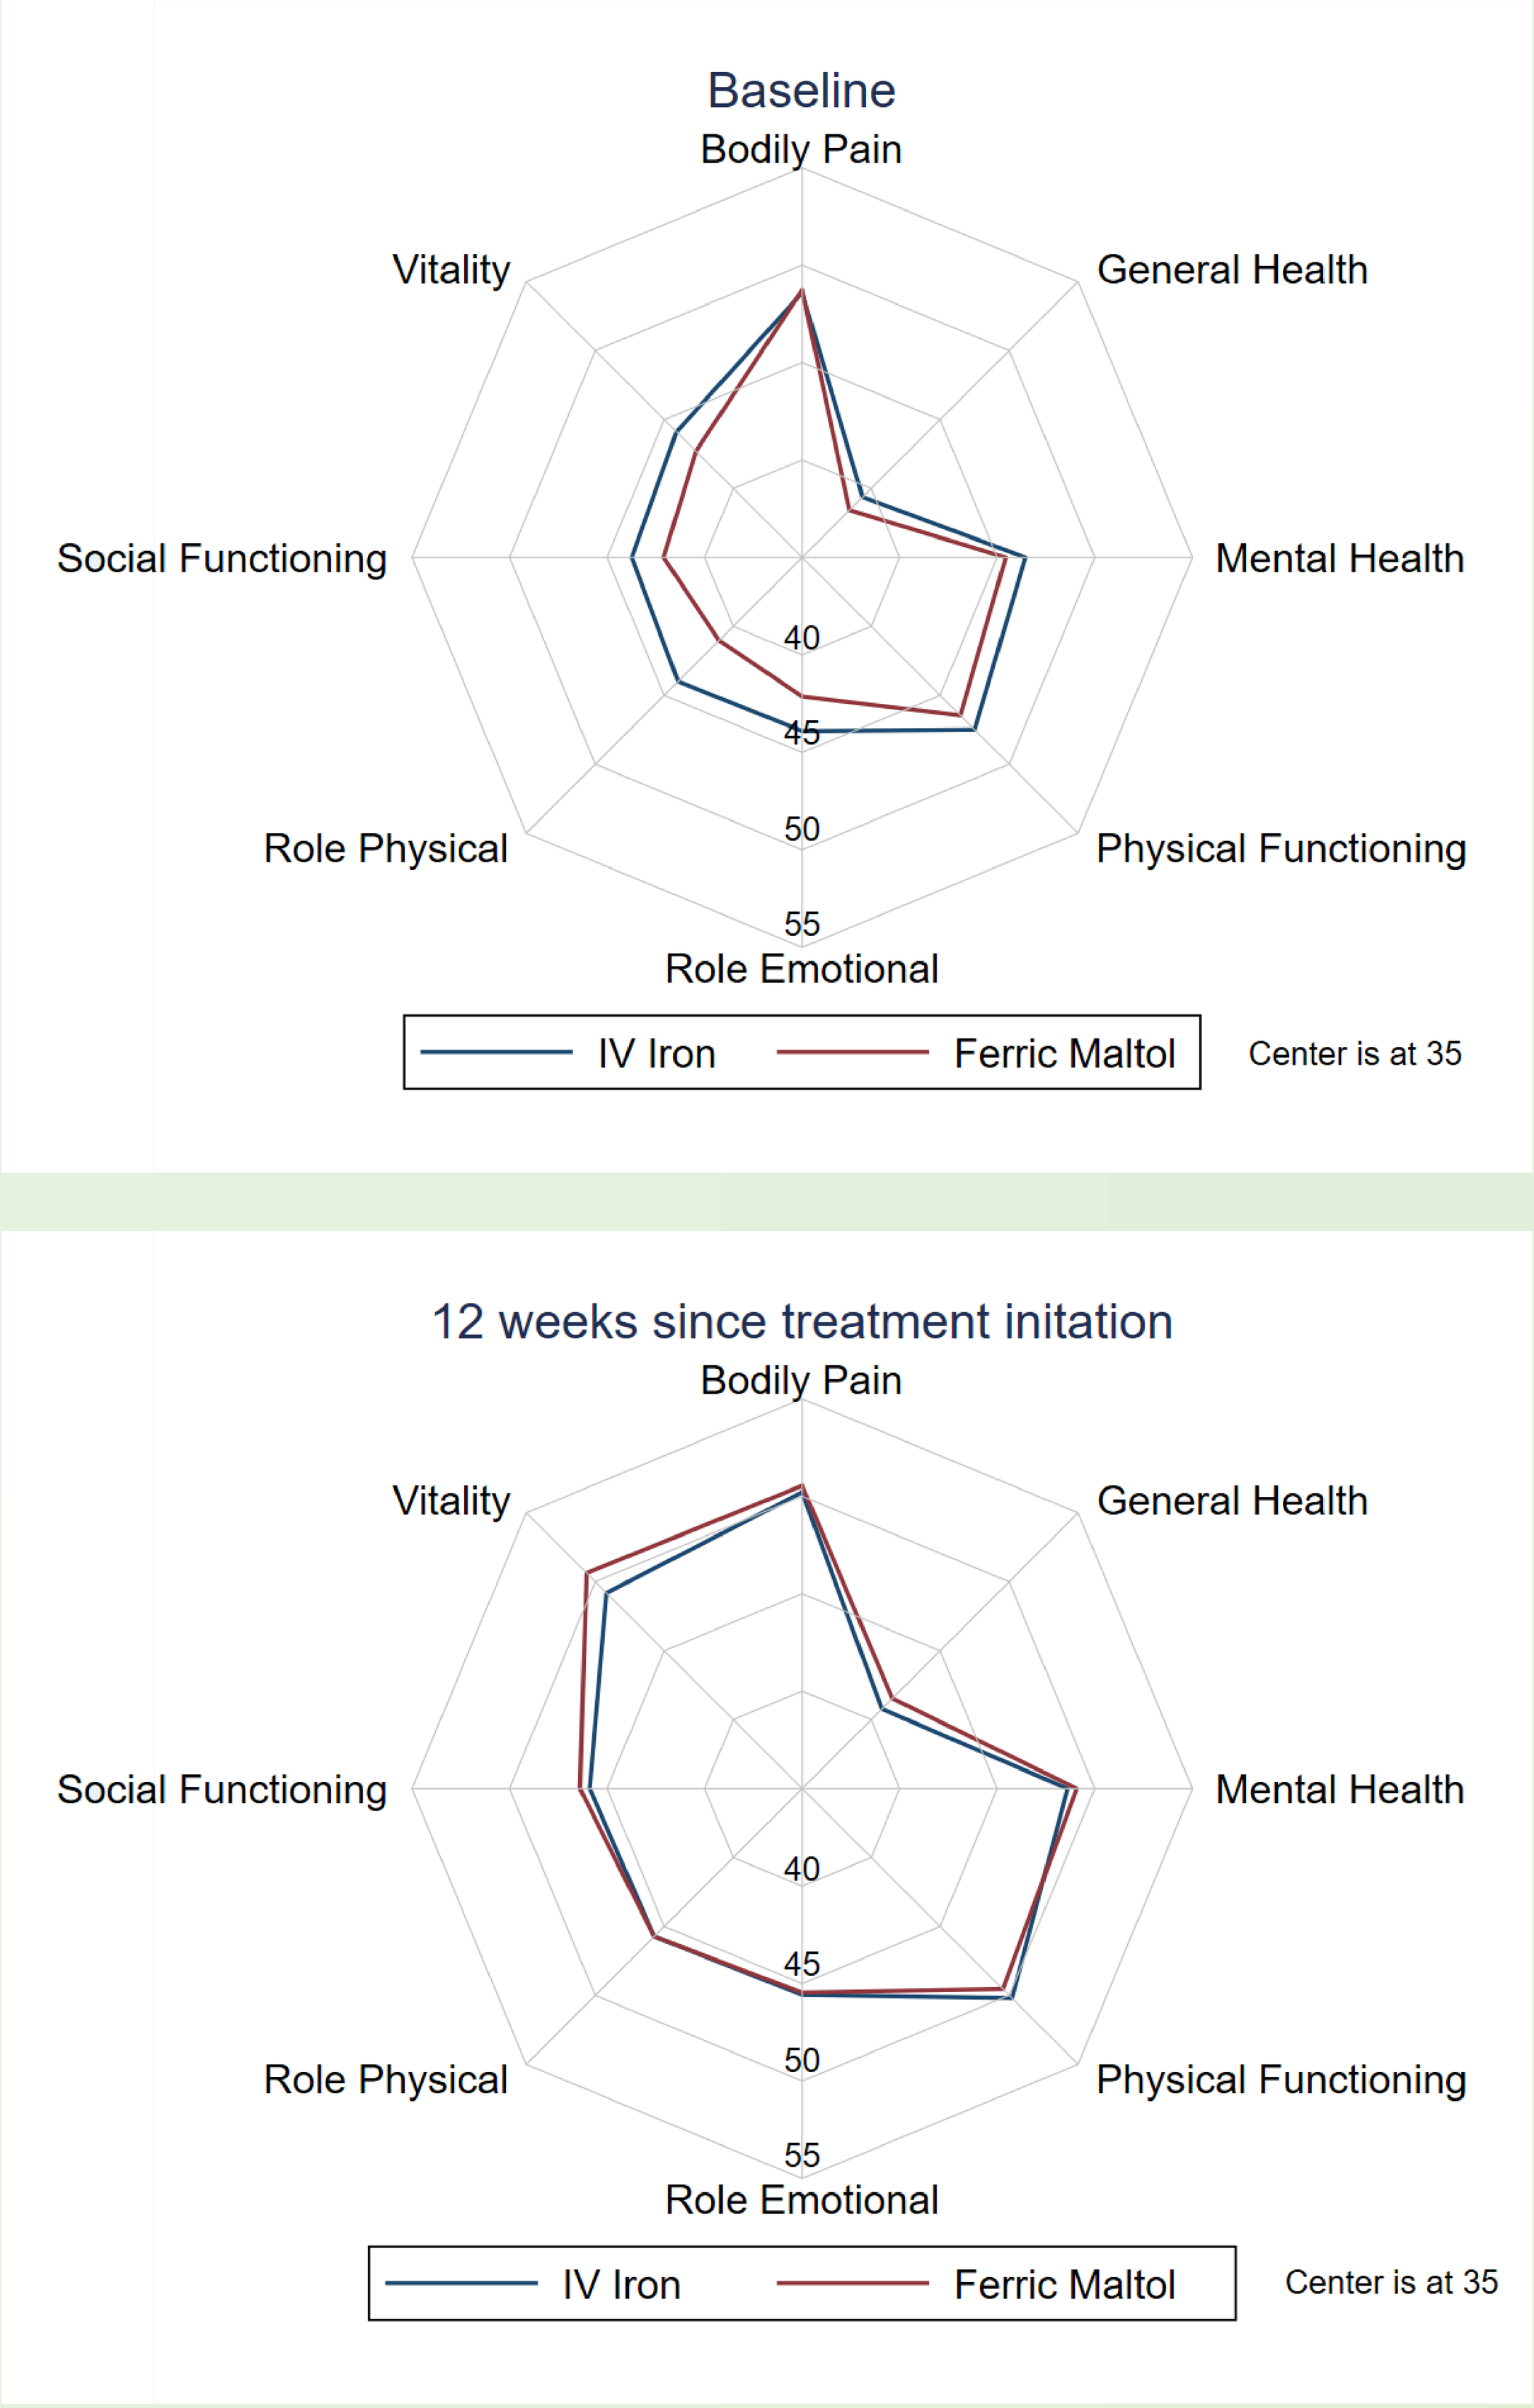

Baseline
Bodily Pain
General Health
Mental Health
Physical Functioning
Role Emotional
Role Physical
Social Functioning
Vitality
40
45
50
55
IV Iron
Ferric Maltol
Center is at 35
12 weeks since treatment initation
Bodily Pain
General Health
Mental Health
Physical Functioning
Role Emotional
Role Physical
Social Functioning
Vitality
40
45
50
55
IV Iron
Ferric Maltol
Center is at 35

Table 2: Correlations between HRQoL (PCS and MCS), Hb and serum iron

	Correlation with Hb*		Correlation with serum iron*	
	PCS	MCS	PCS	MCS
Oral FM and IV FCM	0.227	0.125	0.105	0.091

*Correlations assessed via PCC, FM and IV FCM pooled data

Figure 2: Correlations between Hb and HRQoL (PCS and MCS), and between serum iron and HRQoL (PCS and MCS), assessed at week 12 using PCC

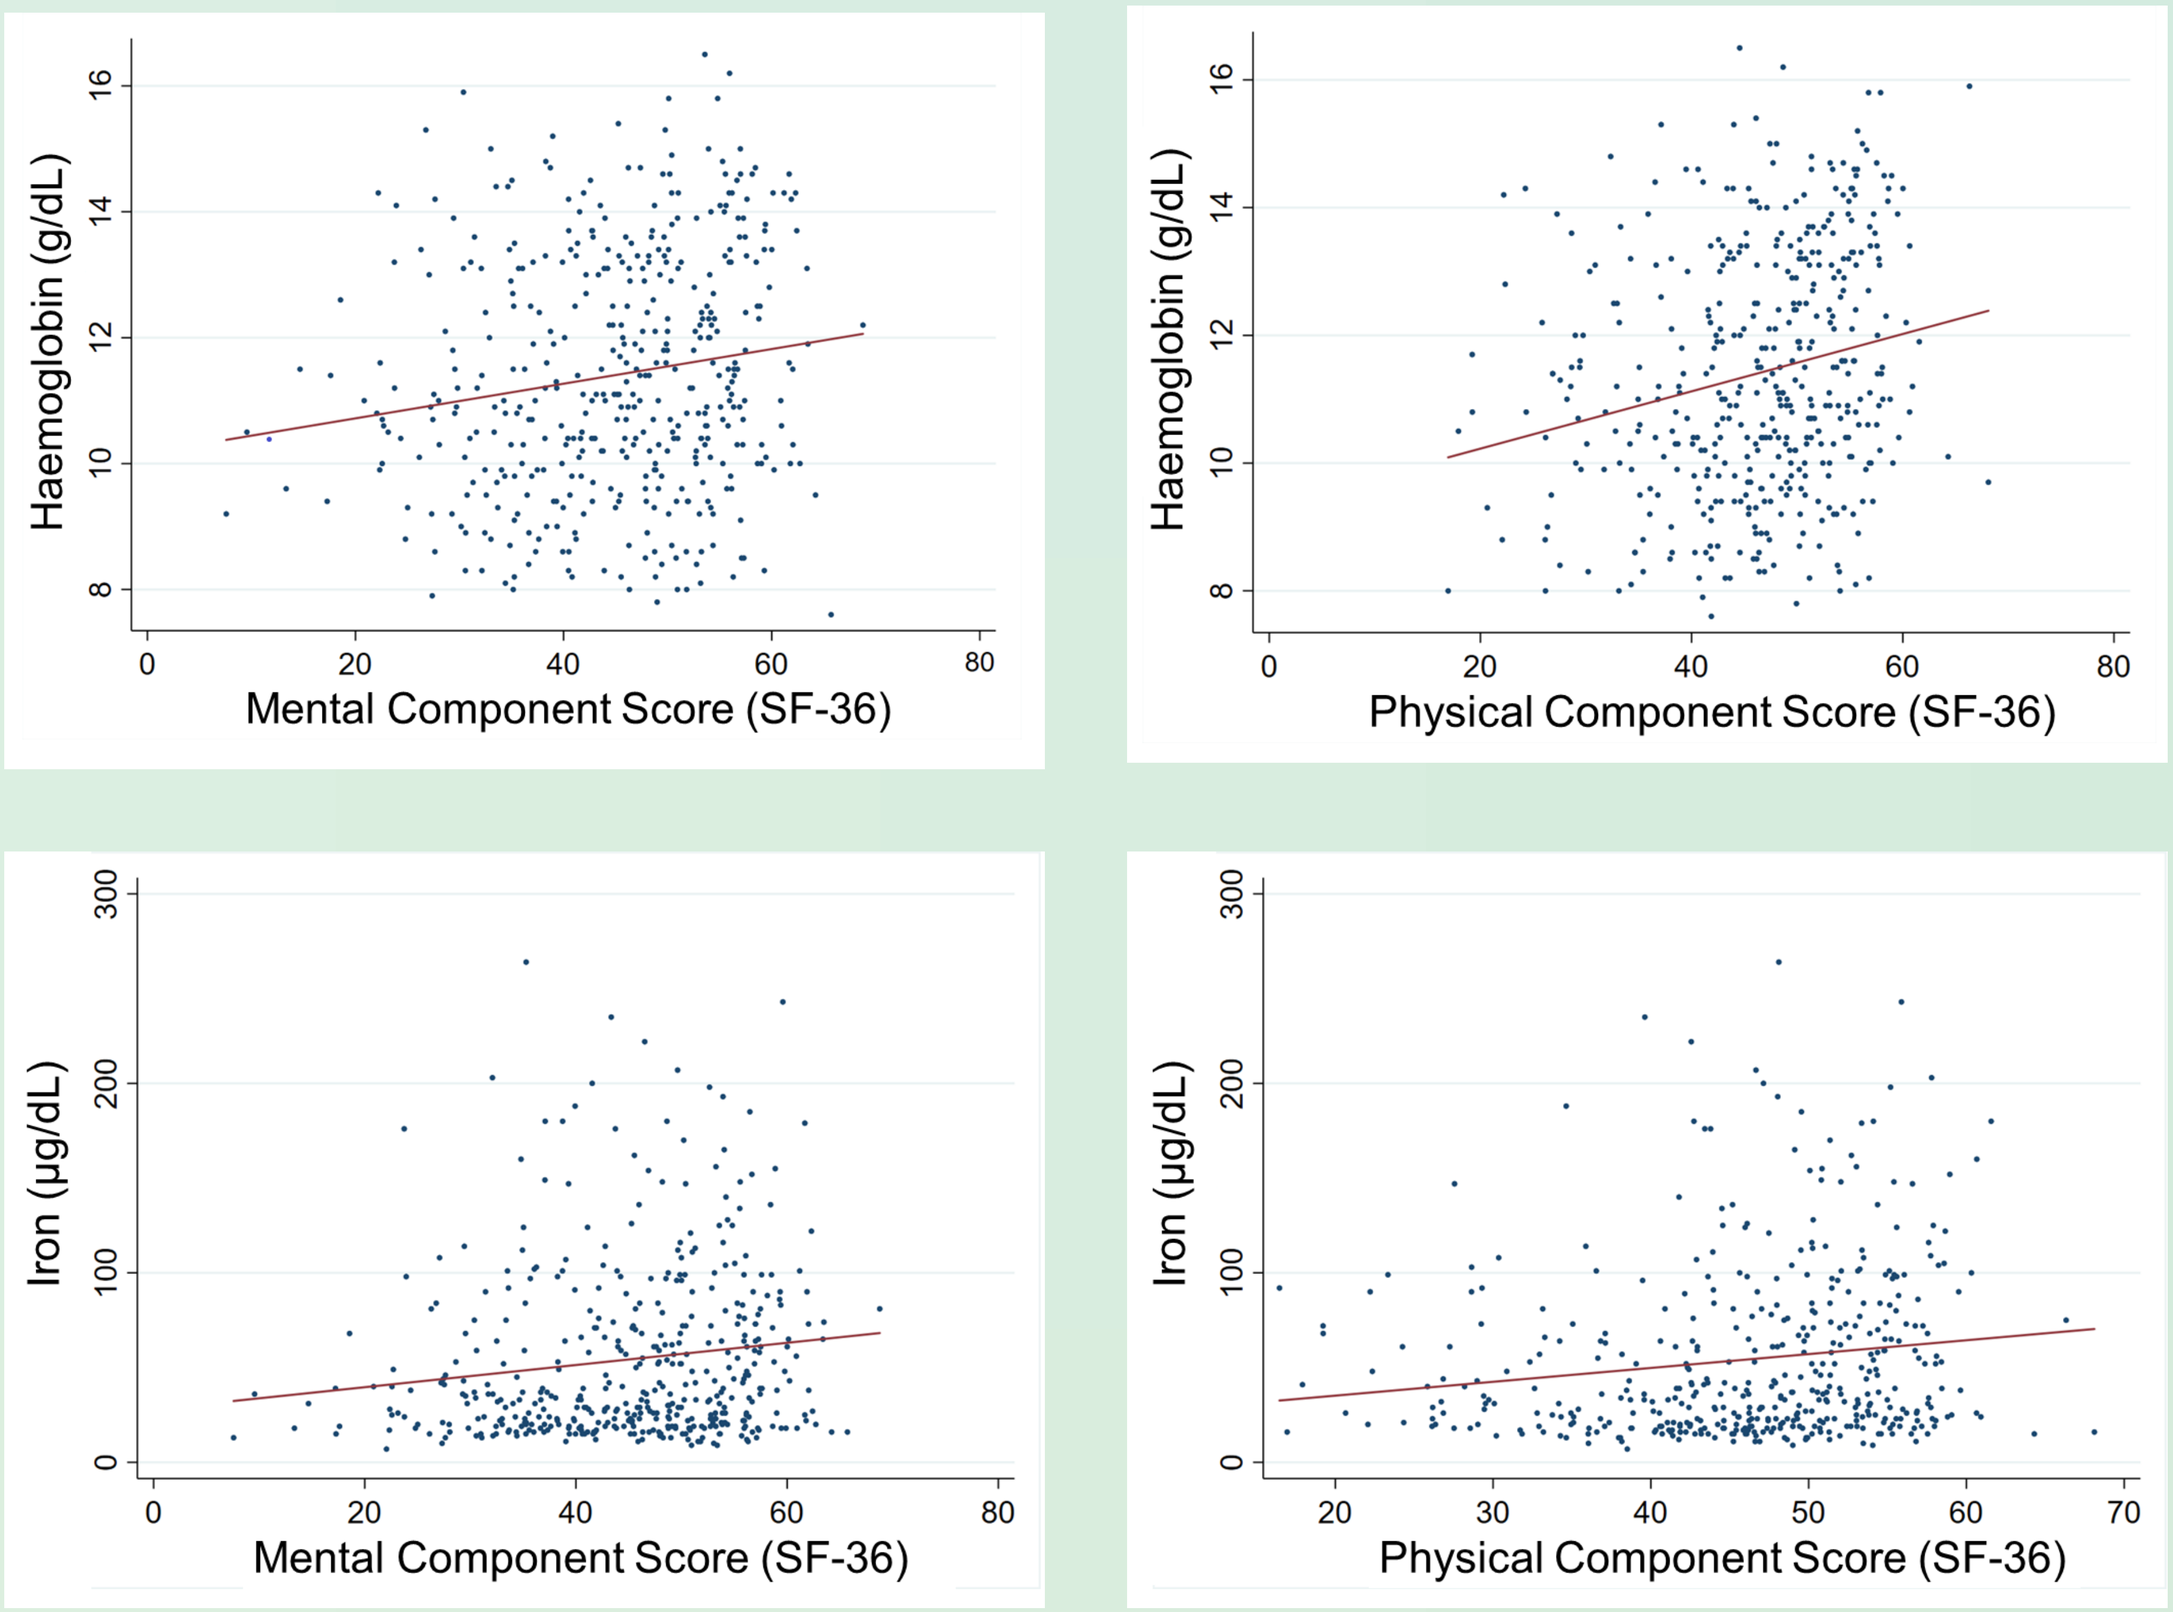

Haemoglobin (g/dL)
Mental Component Score (SF-36)
Haemoglobin (g/dL)
Physical Component Score (SF-36)
Iron (µg/dL)
Mental Component Score (SF-36)
Iron (µg/dL)
Physical Component Score (SF-36)

Conclusions

- **HRQoL as measured by SF-36 PCS and MCS improved following both FM and IV FCM, with no statistically significant difference between treatments. Improvements were positively correlated with increases in Hb and serum iron.**
- **Patient-reported outcomes are an important way to measure health and wellbeing. Measures of HRQoL in IDA have the potential to reflect both IDA symptoms and treatment-related issues such as adverse effects, so are a useful tool to capture additional treatment benefits beyond those typically reported in clinical studies.**
- **HRQoL is thus an important measure of treatment effectiveness when paired with clinical outcomes, from both a patient and decision-making perspective.**
- **FM provides HRQoL benefits at least as great as those with IV FCM, and may provide an oral alternative to IV iron in patients with IBD.**

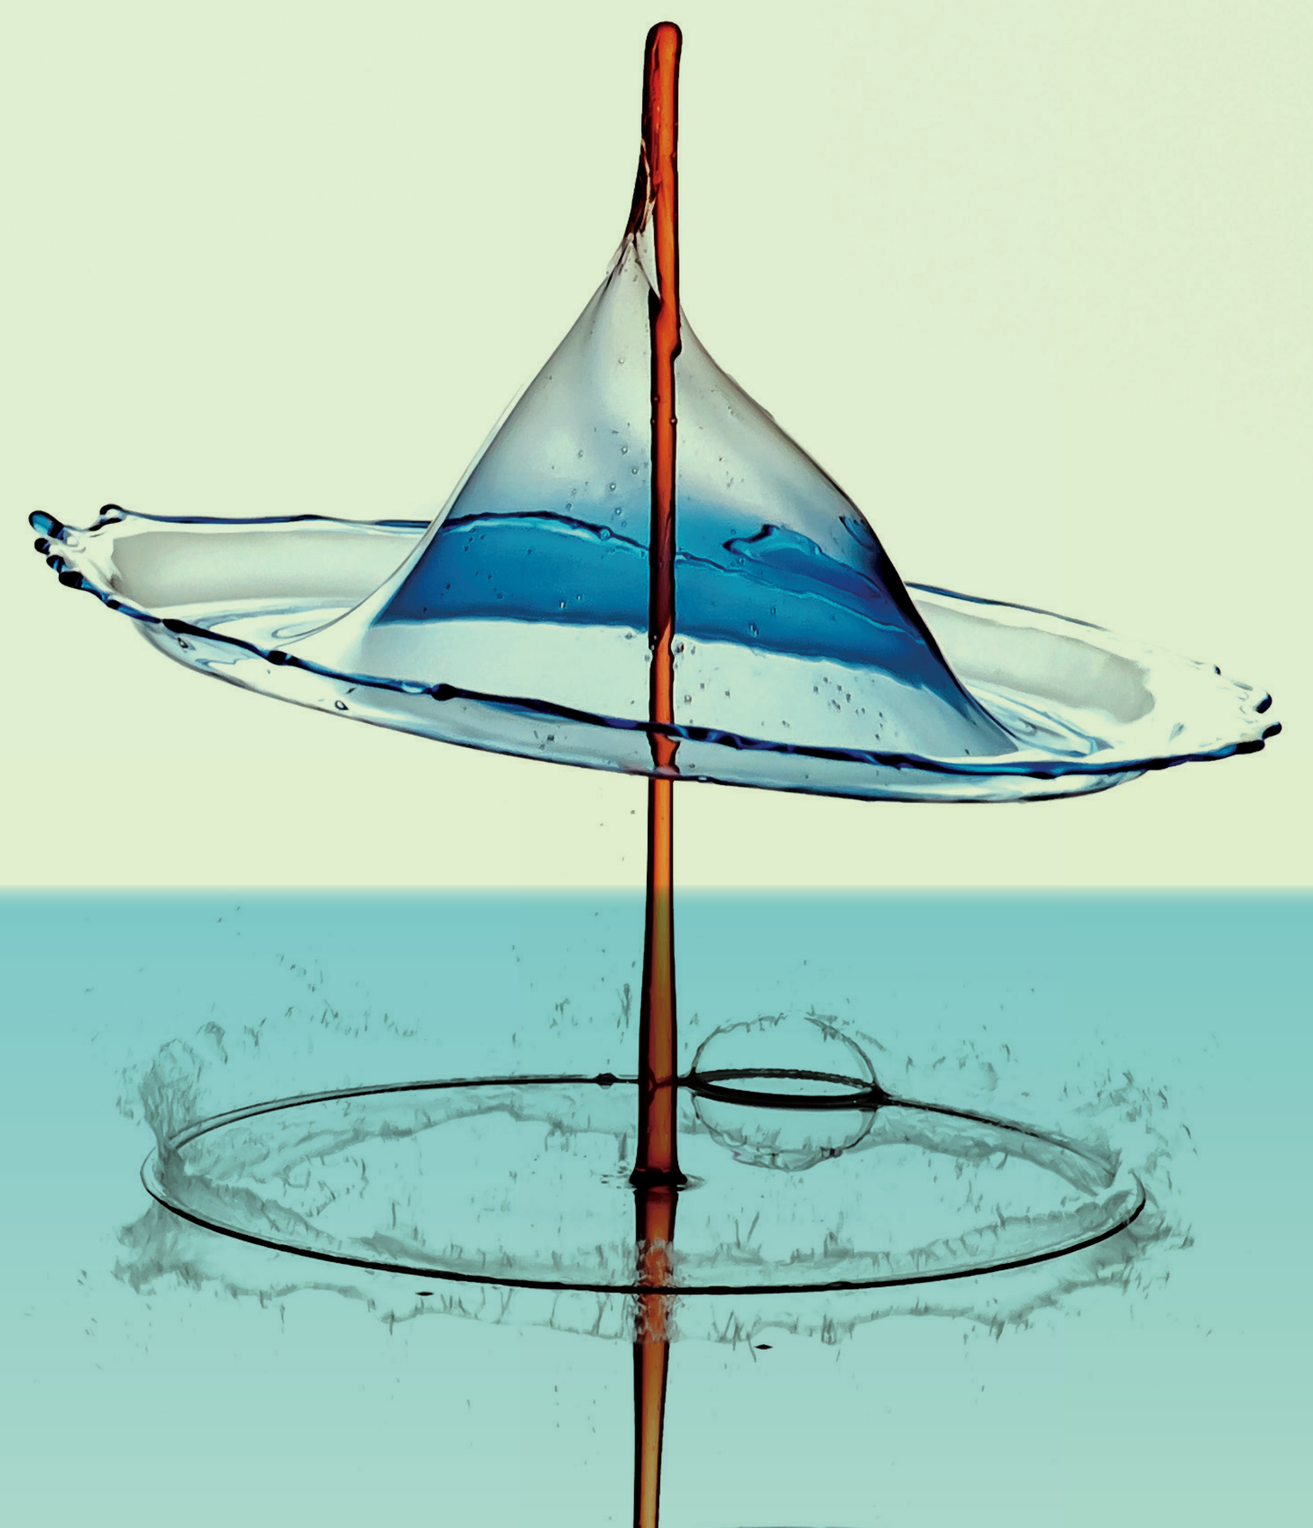

Acknowledgements: The authors acknowledge the contribution of Ben Wilding of HEOR Ltd.

References: 1. Gomollón F et al. World J Gastroenterol. 2009;15(37):4659-65; 2. Stein J et al. Ann Gastroenterol. 2013;26(2):104-13; 3. Peyrin-Biroulet L et al. Am J Clin Nutr. 2015;102:1585-94; 4. Vifor Pharma. Ferinject (ferric carboxymaltose): Summary of Product Characteristics. 2019; 5. Gasche C et al. Inflamm Bowel Dis. 2015;21(3):579-88; 6. Stallmach A et al. Expert Opin Pharmacother. 2015;16(18):2859-67; 7. Norgine Ltd. FERACCRU®: Summary of Product Characteristics. 2019; 8. Howaldt S et al. UEGW 2019, United Eur Gastroenterol J. 2019;7(8 Suppl).

Disclosures: Mark Sampson is an employee and stockholder at Shield Therapeutics PLC. Fatma Akriche is an employee of Norgine Ltd. Ian Jacob is an employee of Health Economics and Outcomes Research Ltd. and received funding from Norgine Ltd. to undertake the analysis outlined in this study. Stefanie Howaldt has nothing to declare. Presented at ECCO (Vienna, Austria) 12-15 February 2020.